Detecting Trace Amounts of Impurity in Methylene Chloride – Sample Preparation Matters

Subhra Bhattacharya, Eric Oliver, Chad Goldman, Apollo Quezada, Mark Jasko, Deva Puranam and Stephen C. Roemer
Thermo Fisher Scientific – Global Chemicals, One Reagent Lane, Fair Lawn, NJ

Abstract

Methylene chloride is widely used as an extraction solvent in pharmaceutical, environmental, food and chemical industries. To achieve the best quality of methylene chloride, trace amounts of impurity must be detected and removed from this solvent. Detection of trace constituents in a solvent can be performed by concentrating a substantial amount of neat solvent in order to gain the desired sensitivity prior to instrumental analysis. Care should be taken during the concentration procedure to ensure the solvent retains all of the constituents. We report a sample preparation technique for methylene chloride wherein the neat sample is spiked with parts per trillion of lindane and recovery of lindane assessed using different concentration techniques such as rotary evaporator, Kurdana-Danish (KD) apparatus, and a combination of both. The KD apparatus showed better recovery compared to the rotary evaporator, however, the most consistent data was observed using a hybrid method of solvent concentration in which a combination of rotary evaporator followed by KD apparatus was used. Trace analysis by GC-ECD showed consistently >98% recovery of lindane using the hybrid method whereas the KD method alone showed only 65% recovery. To our knowledge, this is the first report of a rotary evaporator/KD hybrid method for routine sample preparation and trace analysis in methylene chloride.

Introduction

Chlorinated organic pesticides are widely used against household, garden and plant pest diseases. Some of the pesticides can accumulate in soils, water, fruits and vegetables, and are transported long distances by natural processes. Many of these pesticides can have negative impact on human health. Methylene chloride is a common solvent for the extraction of trace level organic constituents from various sources. Indeed, high purity methylene chloride is used in several EPA methods for the extraction of trace level pesticides, and other contaminated materials from water and soil. Extraction by the solvent followed by evaporation is a common technique for enriching the amount of analyte. Therefore, the solvent needs to be free of any contamination that could interfere with trace level of analyte during extraction. A second concern is retention of the target analyte(s) during the extraction and evaporation processes. To address these issues, trace levels of lindane were added to high purity methylene chloride and spike recovery experiments were carried out using three different evaporation techniques:

- Rotary evaporator (Rota-vap)
- Kurdana-Danish (KD) apparatus
- A combination of both Rota-vap and KD (hybrid)

Materials and Methods

Standards and Solvents

- GC-ECD standard
 - Ultra Scientific Cat. No. US-102BN (2000 µg/ml). Organochlorine pesticide mixture suitable for EPA 508
- GC-MS standard
 - Supelco Supelpreme-HC Internal Standard Mix Cat. No. 4-8902 (2000 µg/ml). The internal standard mix is recommended for EPA 8270 semi-volatile internal standard mix
- Lindane standard (for spiking)
 - Ultra Scientific, Cat. No. EPA-1079
- Solvents
 - Methylene chloride, Fisher Chemical, D151 Optima grade and D154 GC Resolv grade
 - Hexane, Fisher Chemical, H306 Optima grade

Spike Recovery Experiment

- 4L of solvent was spiked with lindane from 10 pg/mL (10 ppt) to 250 pg/mL (250 ppt)
- A standard curve was generated to quantify the amount of lindane from spike recovery experiments
- Spiked samples were concentrated either 500-fold, 1000-fold, or 2000-fold
- Each sample was analyzed at least in triplicate

Solvent Exchange

- Methylene chloride was exchanged with 95% n-Hexane (Fisher Chemical H306 Optima grade) for pesticide testing
- Hexane exchange volume was standardized to get best recovery

Reproducibility of the Assay

- Ultra pure methylene chloride was concentrated using the hybrid method (Rota-vap/KD combination)
- Accuracy and reproducibility of the assay was verified by GC-ECD, GC-FID and GC-MS

Instruments

- Concentrated solvents and analytes were measured using gas chromatography (GC) procedure
- **GC-ECD:** The pesticide residue analysis was performed using gas chromatograph equipped with a Thermo Scientific RSH autosampler, a split-splitless inlet, and an electron capture detector. A 30 m x 0.25 mm x 1.0 micron TR-5 MS capillary column from Thermo Scientific was used for all analyses.
- **GC-MS:** The gas chromatograph was equipped with a Thermo Scientific RSH autosampler, a split-splitless inlet, and ISQ mass detector. A 30 m x 0.25 mm x 0.25 micron TR-1 MS capillary column from Thermo Scientific was used for all analyses.
- **GC-FID:** The gas chromatograph was equipped with an Agilent 6890 system with autosampler, a split-splitless inlet, and flame ionization detector. A 30 m x 0.25 mm x 1.0 micron TR-5 MS capillary column from Thermo Scientific was used for all analyses

GC-ECD Run Conditions

- Column: Thermo Scientific TR-1 Gold (or equivalent)
- Column Dimension: 30 Meter x 0.25 mm x 1 micron
- Column flow: 2 mL/min (constant flow mode)
- Carrier Gas: Helium
- Oven Temp: 40 deg C initial
- Ramp rate: 15 deg C/min, final temp 250 deg C, hold: 15 min
- Injection Temp: 220 deg C
- Injection Volume: 1 µl
- Injection mode: Splitless
- Split Flow: 50 mL/min after 1 min

Results

- Comparison of spike recovery using the KD and Rotavap concentration method (Table 1) and the spike recovery result from hybrid method (Table 2) clearly indicates better recovery by the hybrid method.
- Amount of solvent draw-down during evaporation (Table 2) plays a key role in the analyte recovery. More than 98% recovery was observed in GC-ECD when 1000 mL of spiked solvent was reduced to 1 mL compared to 86% recovery when concentrating to 0.5 mL
- Greater than 98% recovery was consistently obtained using the preferred Rota-vap/KD concentration method including the hexane exchange (Table 2 and Figs. 1, 2)
- Ultrapure solvent from two different bottles was concentrated using the hybrid method to ensure the reproducibility of the hybrid method (Fig. 1). Solvent peaks are compared from the results generated by two different chemists.
- Ultrapure $MeCl_2$ with 1000:1 hybrid concentration showed no GC-ECD pesticide peaks in the 8-25 min RT relative to the EPA 508 pesticide standard (Fig. 2)
- Ultrapure $MeCl_2$ with 1000:1 hybrid concentration showed no GC-FID impurity peaks in the 7-21 min RT (Fig. 3)
- Ultrapure $MeCl_2$ with 1000:1 hybrid concentration showed no GC-MS impurity peaks in the 7-20 min RT relative to the 6-point GC-MS standard (Table 3 and Fig. 4)

Table 1. Percent Recovery (by GC-ECD) of Lindane Spiked in $MeCl_2$ and Concentrated Using KD Apparatus or Rotary Evaporator Alone

Sample #	Spike Recovery (%) Using KD	Spike Recovery (%) Using Rota-Vap
1	61.92	27.28
2	58.76	28.49
3	63.13	33.29
4	58.72	29.69
5	66.20	27.97
6	73.64	26.21
7	72.85	37.88
Mean	**65.03**	**30.12**
Standard Deviation	**6.18**	**4.10**

Comparison of GC-ECD data after spike recovery.

Table 2. Percent Recovery (by GC-ECD) of Lindane Spiked in $MeCl_2$ and Concentrated Using Rotavap-KD Hybrid Method

	Recovery Amount (ppb)	% Recovery	Recovery Amount (ppb)	% Recovery
Sample #	1000 mL to 0.5 mL Concentration		1000 mL to 1 mL Concentration	
1	17.993	89.97	9.89	98.88
2	17.919	89.55	9.98	99.84
3	15.772	78.86	9.88	98.75
Mean		**86.13**		**99.16**
Standard Deviation		**6.3**		**0.6**

In both experiments, 2 x 10 mL hexane exchange was performed.
Rotavap/KD hybrid with a 1000:1 concentration is the preferred sample preparation method.

Figure 1. Reproducibility of Solvent Peaks Concentrated by Hybrid Method (1000 to 1) Using GC-ECD

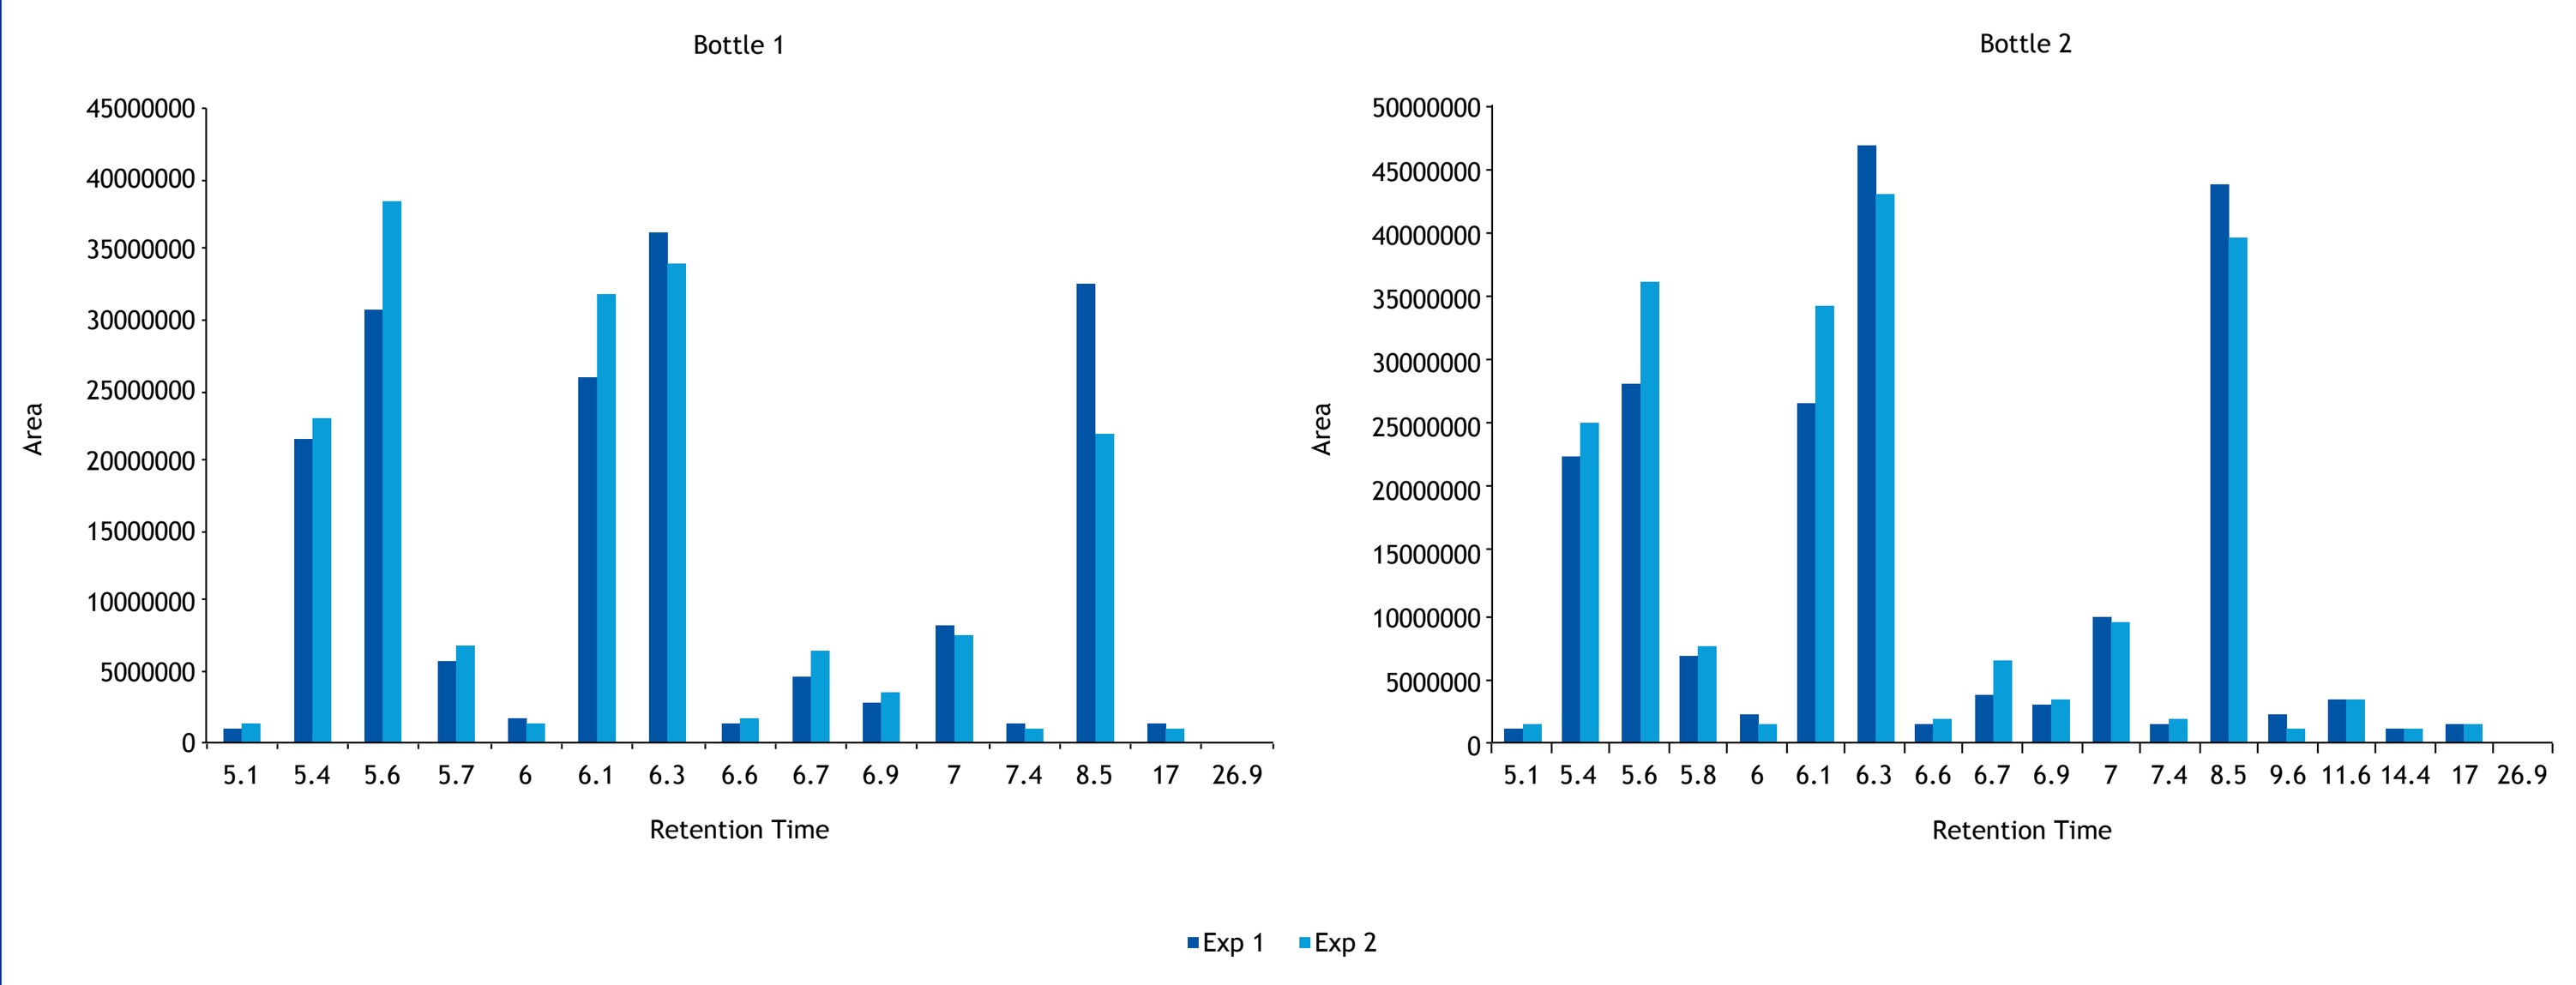
Bottle 1
Bottle 2
Area
Retention Time
Exp 1
Exp 2

Figure 2. Ultrapure $MeCl_2$ Concentrated by Hybrid Method (1000 to 1) and Compared With EPA 508 Standard Using GC-ECD

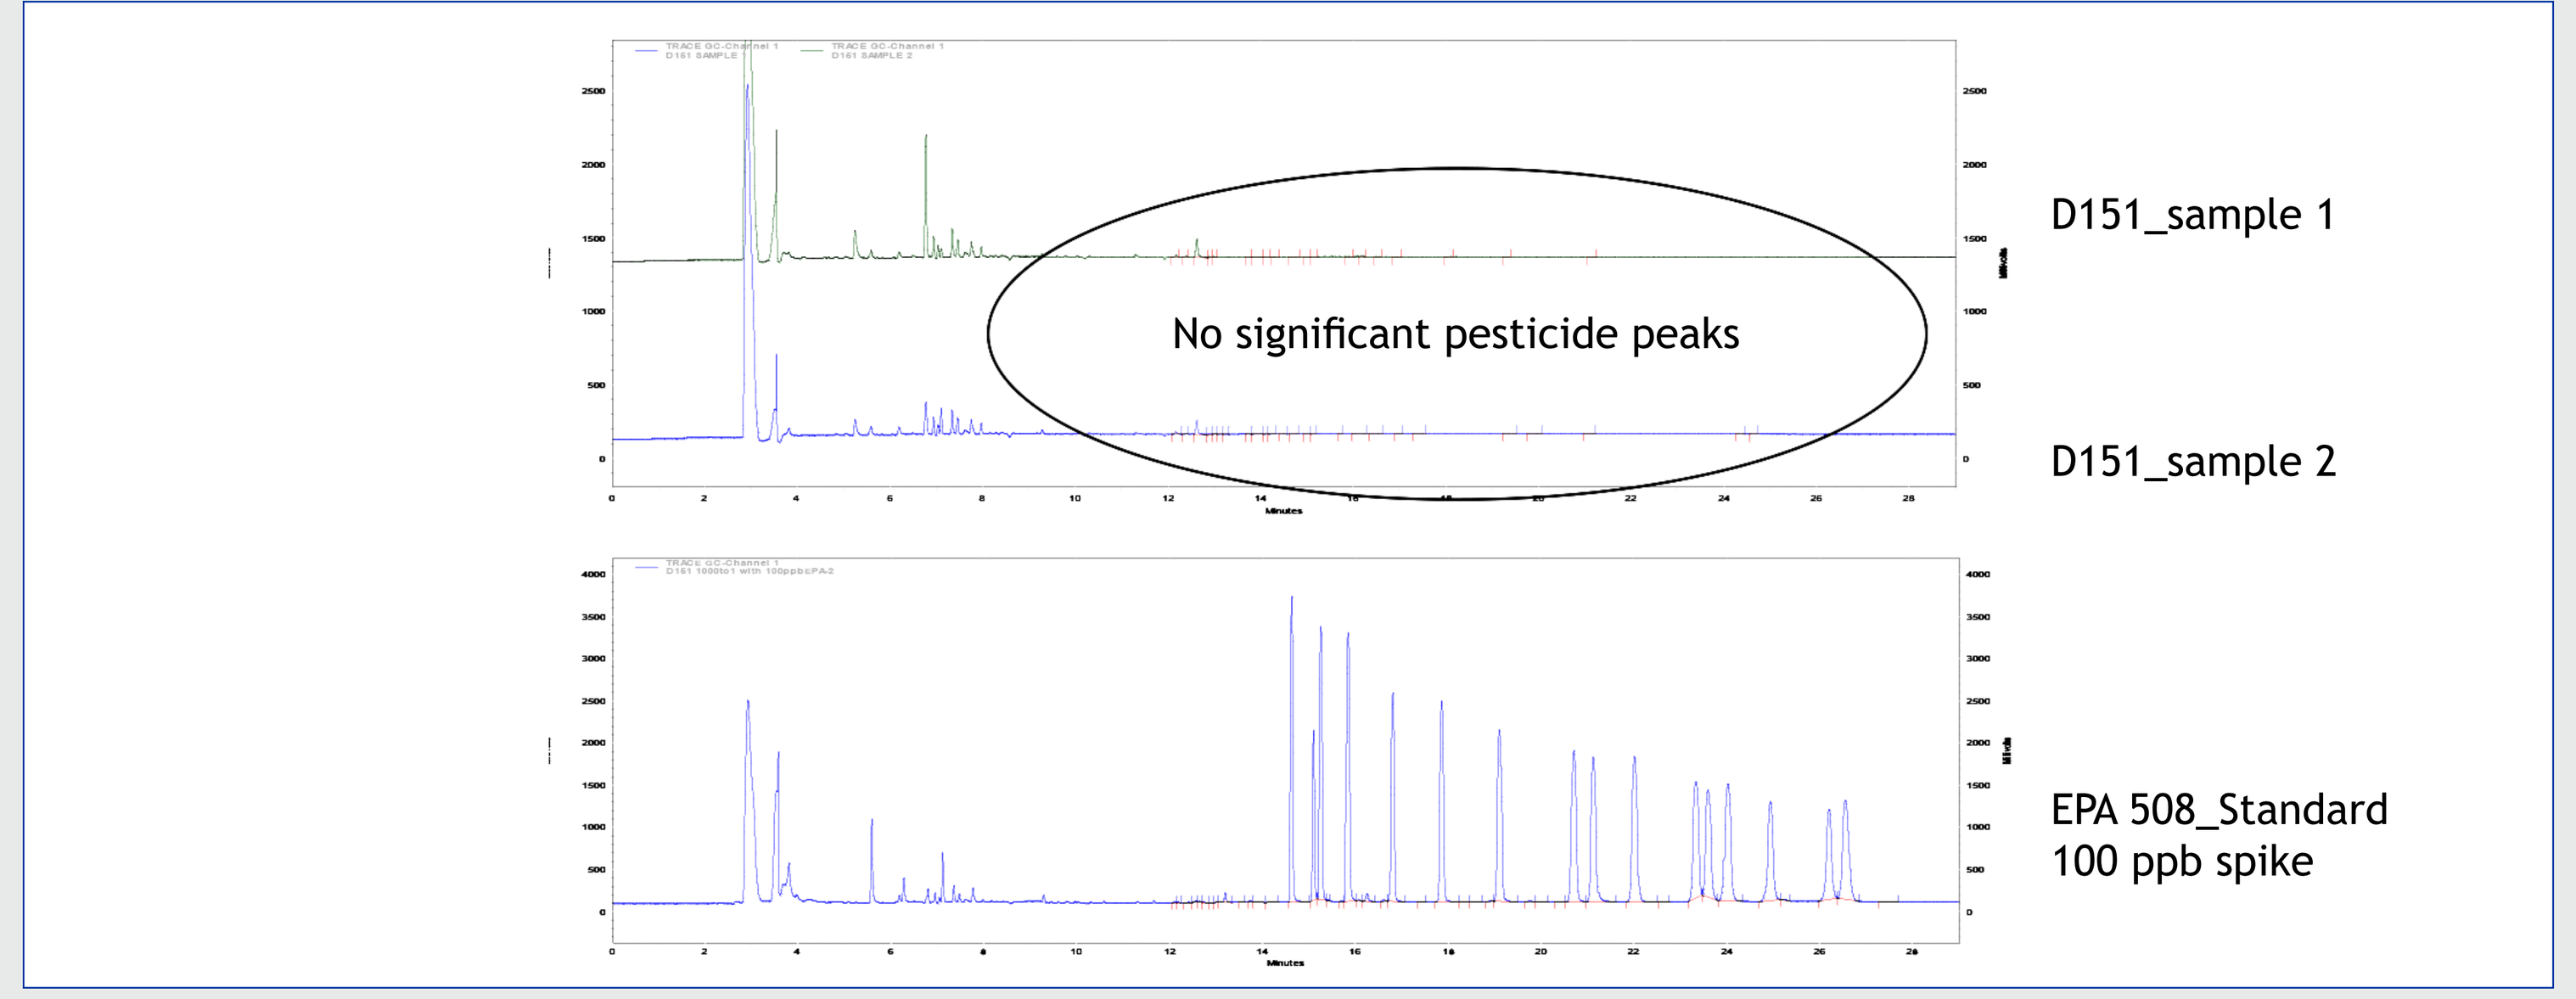
No significant pesticide peaks
D151_sample 1
D151_sample 2
EPA 508_Standard
100 ppb spike

Figure 3. Ultrapure $MeCl_2$ Concentrated by Hybrid Method (1000 to 1) and Analyzed by GC-FID

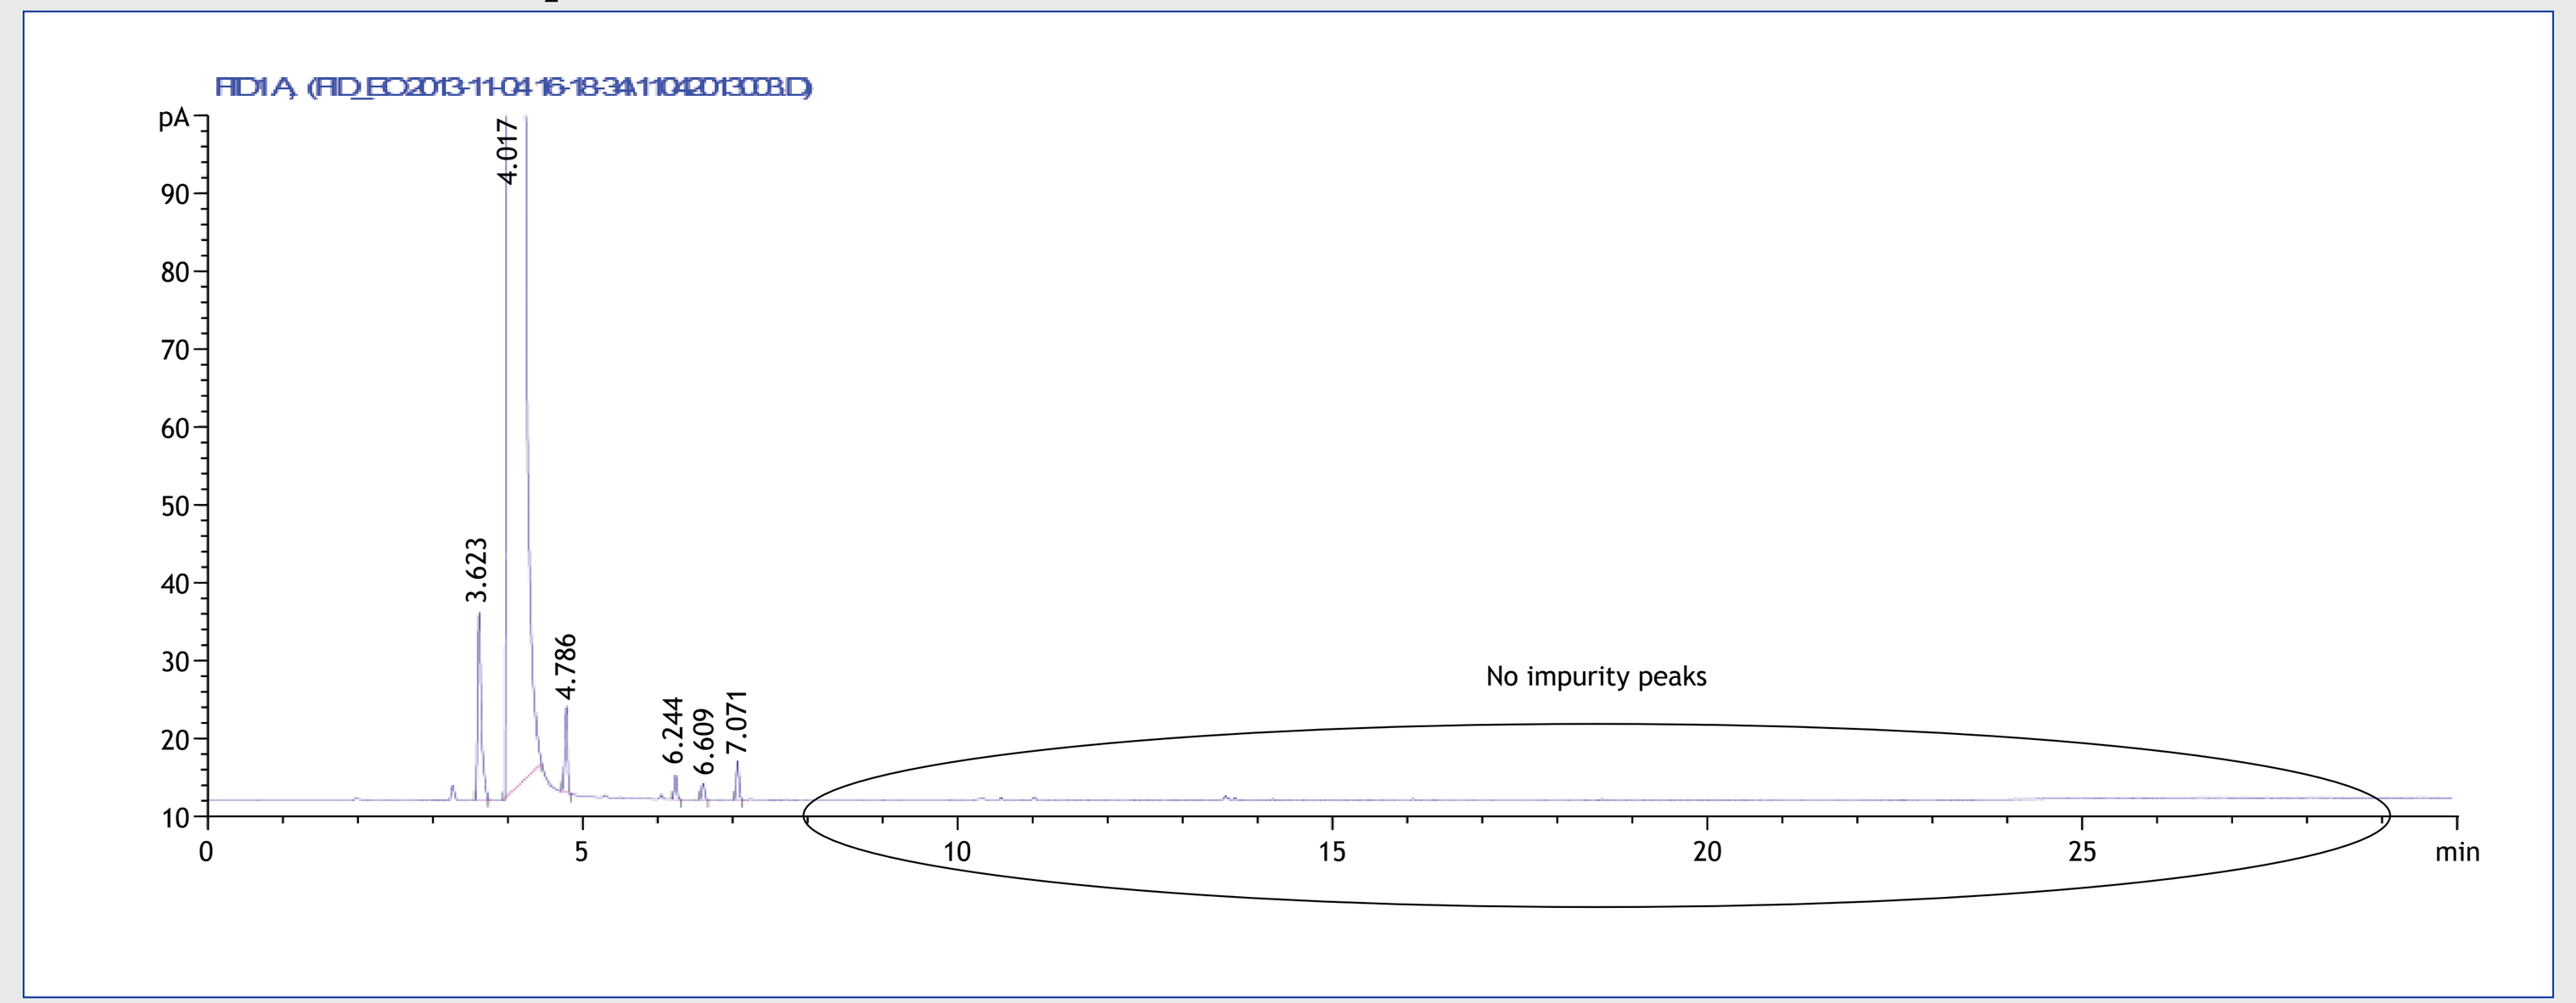
pA
3.623
4.017
4.786
6.244
6.609
7.071
No impurity peaks
min

Table 3. GC-MS Standard Was Run at 4 ppm Concentration. The Six Compounds in the Standard Were as Follows:

Internal Standard RT (min)	Mass (m/z)	Compound
~ 8.13	150.07	D4 — 1,4-dichlorobenzene
~ 9.69	136.2	D8 — Naphthalene
~ 11.81	164.21	D10 — Acenaphthene
~ 13.55	188.22	D10 — Phenanthrene
~ 16.80	240.12	D12 — Chrysene
~ 19.52	264.26	D12 — Perylene

Figure 4. Ultrapure $MeCl_2$ Concentrated by Hybrid Method (1000 to 1) and Analyzed by GC-MS

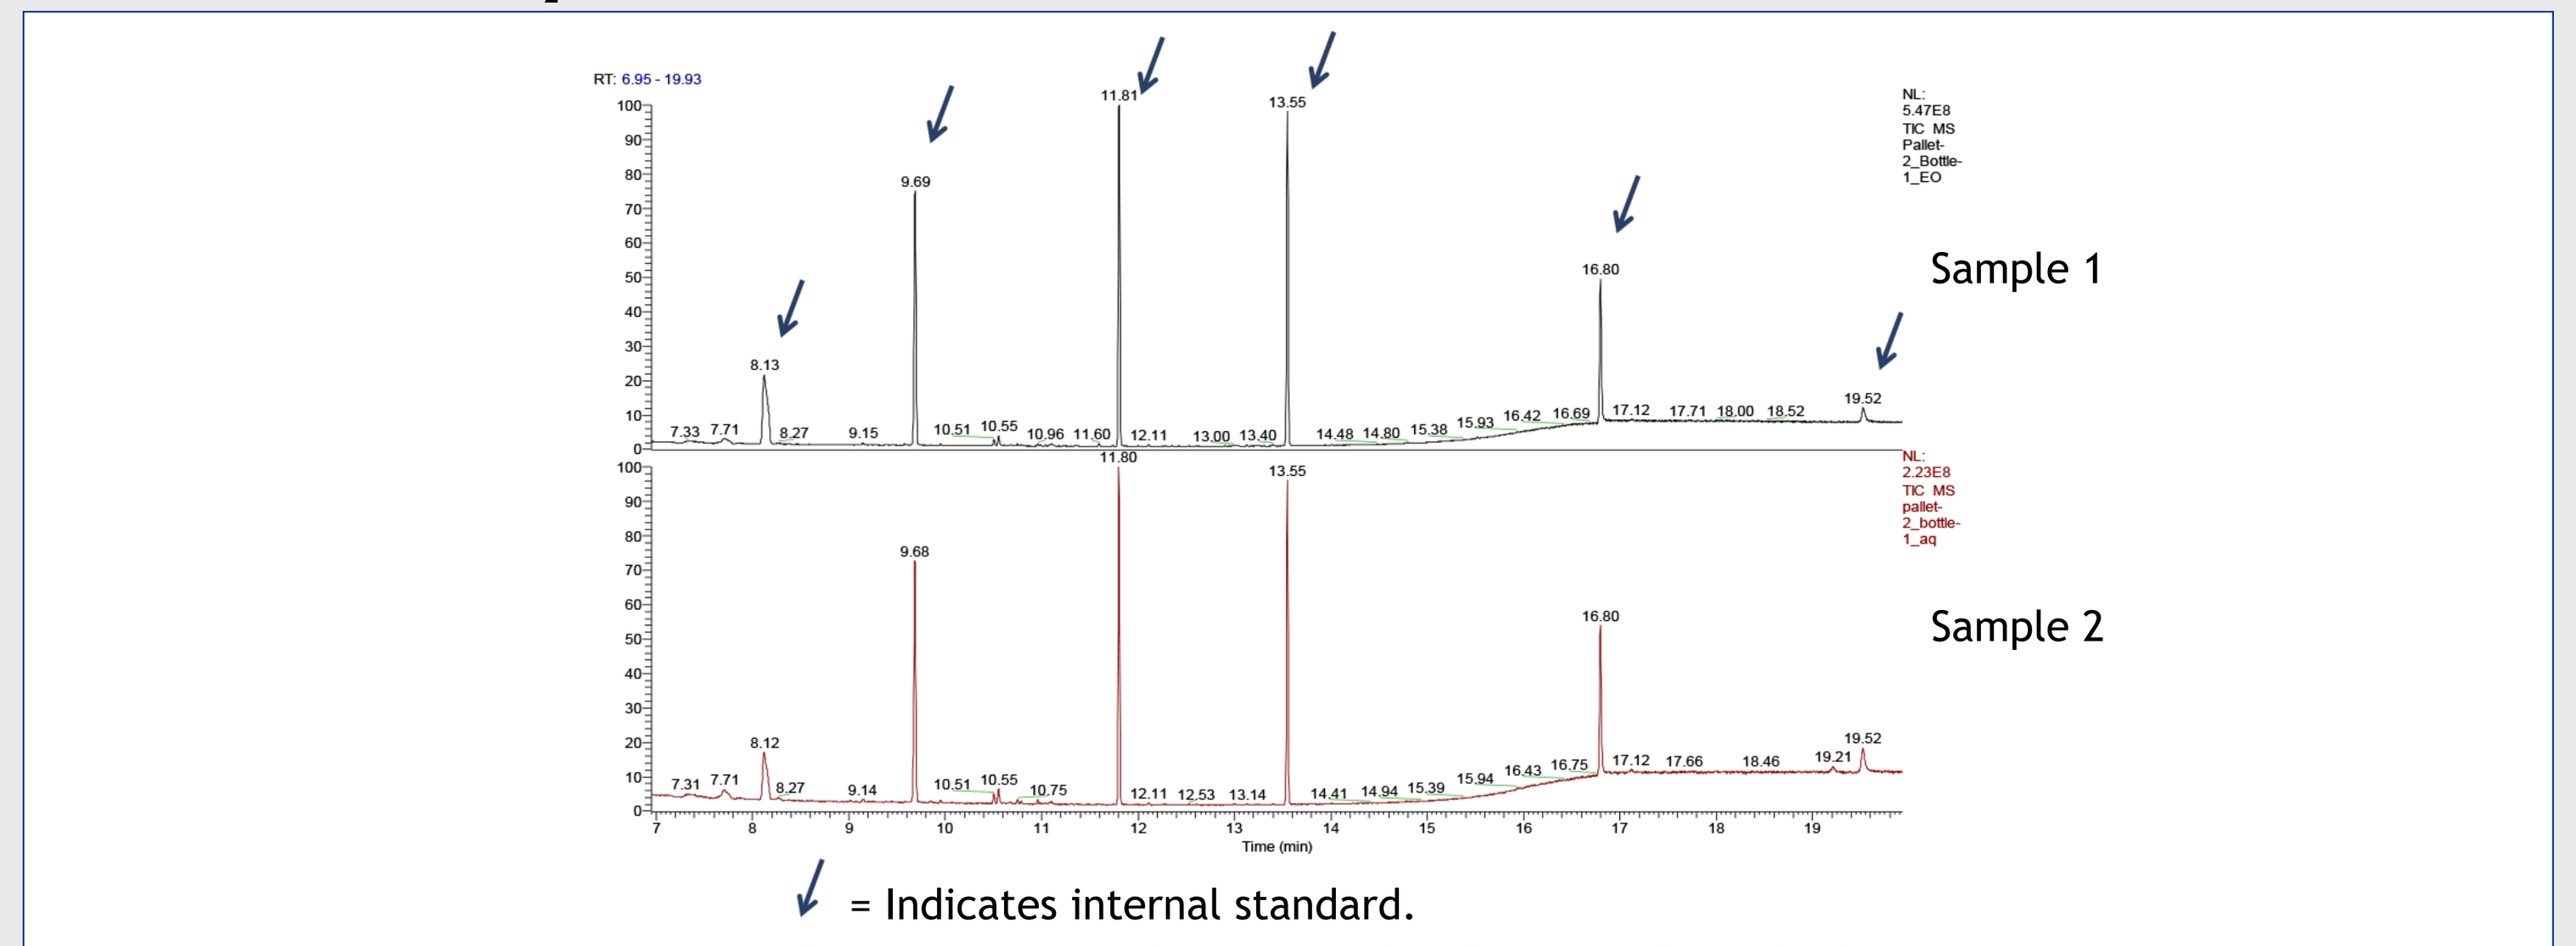
Sample 1
Sample 2
Time (min)

= Indicates internal standard.
No significant impurity peaks compared to the internal standards.

Discussion

- Lindane was chosen as the analyte for spike recovery since poor recovery of this analyte was reported.[1] We observed very similar results when the spiked solvent was concentrated using rotary evaporator. The analyte loss may be associated with flushing of solute layers from dry surfaces (co-evaporation) during sample evaporation.[2]
- Even though concentration of the spiked solvent using KD apparatus showed better recovery, reproducibility is not very consistent.
- With respect to the hybrid method of Rota-vap and KD, greater than 98% analyte recovery with high reproducibility was obtained as noted by GC-ECD. Amount of solvent evaporated plays a key role in the recovery of analyte. Amount of hexane is another key factor for the recovery of analyte. We observed 2 x 10 mL hexane exchange works best compared to other amounts (data not shown).
- A significant amount of time is saved by hybrid method (45 min) compared to KD method alone (2 hr).
- Reproducibility testing was accomplished by concentration of the neat solvent. The goal of the experiment was to show that Fisher Chemcial methylene chloride (D151 and D154) is ultrapure and can be used for trace level (ppt) detection of target analytes without any interference from the solvent. We achieved this goal as evidenced by GC-ECD, GC-FID, and GC-MS chromatograms.
- Our data showed there are no interfering peaks of any semi-volatile pesticide residues as evidenced by chromatograms from FID, ECD and mass spectrometer.

Conclusions

- Rota-vap/KD hybrid method of sample preparation showed consistent analyte recovery and reproducibility.
- Consistent assay reproducibility observed by GC-MS, GC-ECD and GC-FID.
- GC-ECD, GC-FID and GC-MS data showed interference-free baseline for ultrapure methylene chloride (Fisher Chemical D151 and D154) which indicated these are the solvents of choice for today's advanced instrument systems.

References

1. C.C. Cheng., *Polycyclic Aromatic Compounds*, 23, 315–325 (2003).
2. M.D. Erickson, M.T. Giguere, and D.A. Whitaker., *Analytical Letters*, 14(11), 841–857 (1981).

Fisher Chemical

Fisher Scientific
Part of Thermo Fisher Scientific